RHESUS ANTIBODY SECRETING CELLS DIFFERENTIATED *EX VIVO* FROM B CELLS ENGRAFT WITHOUT PRECONDITIONING IN AN AUTOLOGOUS HOST AND REPRESENT A NOVEL MODALITY FOR CELL AND GENE THERAPY

David J Young[1], Jacob Tuner[2], Abigail Edwards[2], Ella Liberzon[2], Sogun Hong[1], Noriko Sato[3], Adam S Lazorchak[2], Richard A Morgan[2], Cynthia E Dunbar[1]

1. National Heart, Lung, and Blood Institute, National Institutes of Health, Bethesda, MD 20892
2. Be Biopharma, Cambridge, MA 02139
3. National Cancer Institute, National Institutes of Health, Bethesda, MD 20892

Manufactured cell therapies have produced transformative medicines for previously intractable diseases. Yet barriers limit broader application including insufficient biosynthetic capacity, durability and a need for toxic conditioning to favor engraftment. Terminally differentiated plasma cells (PC) offer natural longevity (mean half-life of 17 years), high levels of protein secretion (10,000 molecules/cell/sec) and resistance to allogeneic rejection, making them attractive for delivering biologic therapeutics that require continuous exposure for therapeutic benefit. Key to developing PCs derived from genetically engineered B cells (B Cell Medicines, BeCM) is preclinical testing in a model with an intact immune system similar to that of humans. Here we present an *ex vivo* method to engineer, expand and differentiate non-human primate (NHP) peripheral blood B lymphocytes into antibody secreting cells (ASC). We show these cells home to and engraft in the bone marrow (BM), liver and spleen of an unconditioned, immune competent host.

We apheresed rhesus macaques (RM) and enriched peripheral blood B lymphocytes by positive selection to generate working B cell banks. We expanded B cells over 8 days in defined serum-free culture to yield ≥10-fold expansion with a final viability ≥80%. ASC production was demonstrated by Ig class switch (70%) with >25% of cells expressing markers consistent with NHP PCs (CD31 and CD138). ELISPOT confirmed IgG, IgM and IgA secretion.

We used zirconium-89-oxine cell radiolabeling for high sensitivity PET/CT tracking of *ex vivo* expanded and differentiated autologous PCs in unconditioned NHPs. We intravenously infused labeled PCs (80 million cells/4.8 MBq) from day-8 cultures. Serial PET/CT images were acquired at 15 minutes, 1 hour and then 1, 3 and 6 days post-infusion. PCs first accumulated in the lungs, shifting to the spleen, liver and BM by 1 hour. Cells left the lung by day 1, reaching steady state in BM, liver and spleen, persisting for at least 6 days. This is consistent with expected homing patterns of ASCs including long-lived PCs.

To produce BeCMs, we developed methodology to engineer NHP B cells using CRISPR/Cas9 gene editing with AAV-mediated homology directed DNA repair (HDR). CRISPR/Cas9 gene editing of NHP B cells at the *CCR5* safe-harbor locus (>70%) was achieved by RNP electroporation. AAV-

mediated HDR knock-in of the NHP sodium-iodide symporter (NIS) into *CCR5* results in transgene expression in the day-8 B cell product. These engineered cells will enable long-term tracking of *NIS* expressing BeCMs *in vivo* by PET/CT imaging in immune competent, autologous NHPs.

In summary, we present a method for differentiating antibody secreting NHP PCs from peripheral blood B cells. We demonstrate short-term homing and engraftment in an autologous, immune competent unconditioned host. Engraftment without preconditioning broadens BeCM clinical utility for patients where preconditioning toxicities are unacceptable or outweigh therapeutic benefit. The successful expression of reporter genes in NHP PCs will allow long-term NHP engraftment studies to address translationally relevant questions such as engraftment durability, reinfusion and long-term biodistribution of BeCMs.

Representative immunophenotyping and imaging of ^{89}Zr-labeled plasma cells

A. Pre-infusion phenotyping

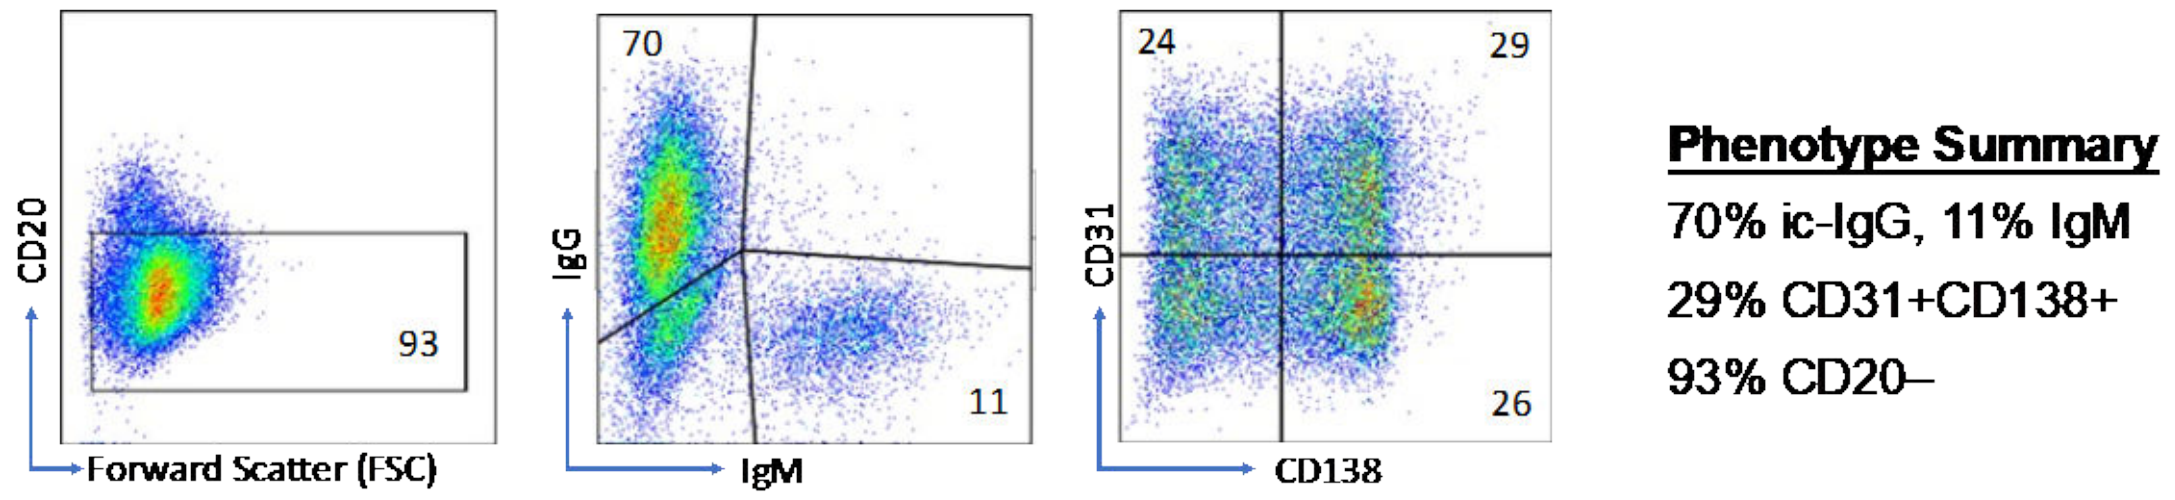
CD20
93
Forward Scatter (FSC)
IgG
70
11
IgM
CD31
24
29
26
CD138
Phenotype Summary
70% ic-IgG, 11% IgM
29% CD31+CD138+
93% CD20–

B. Serial PET/CT imaging

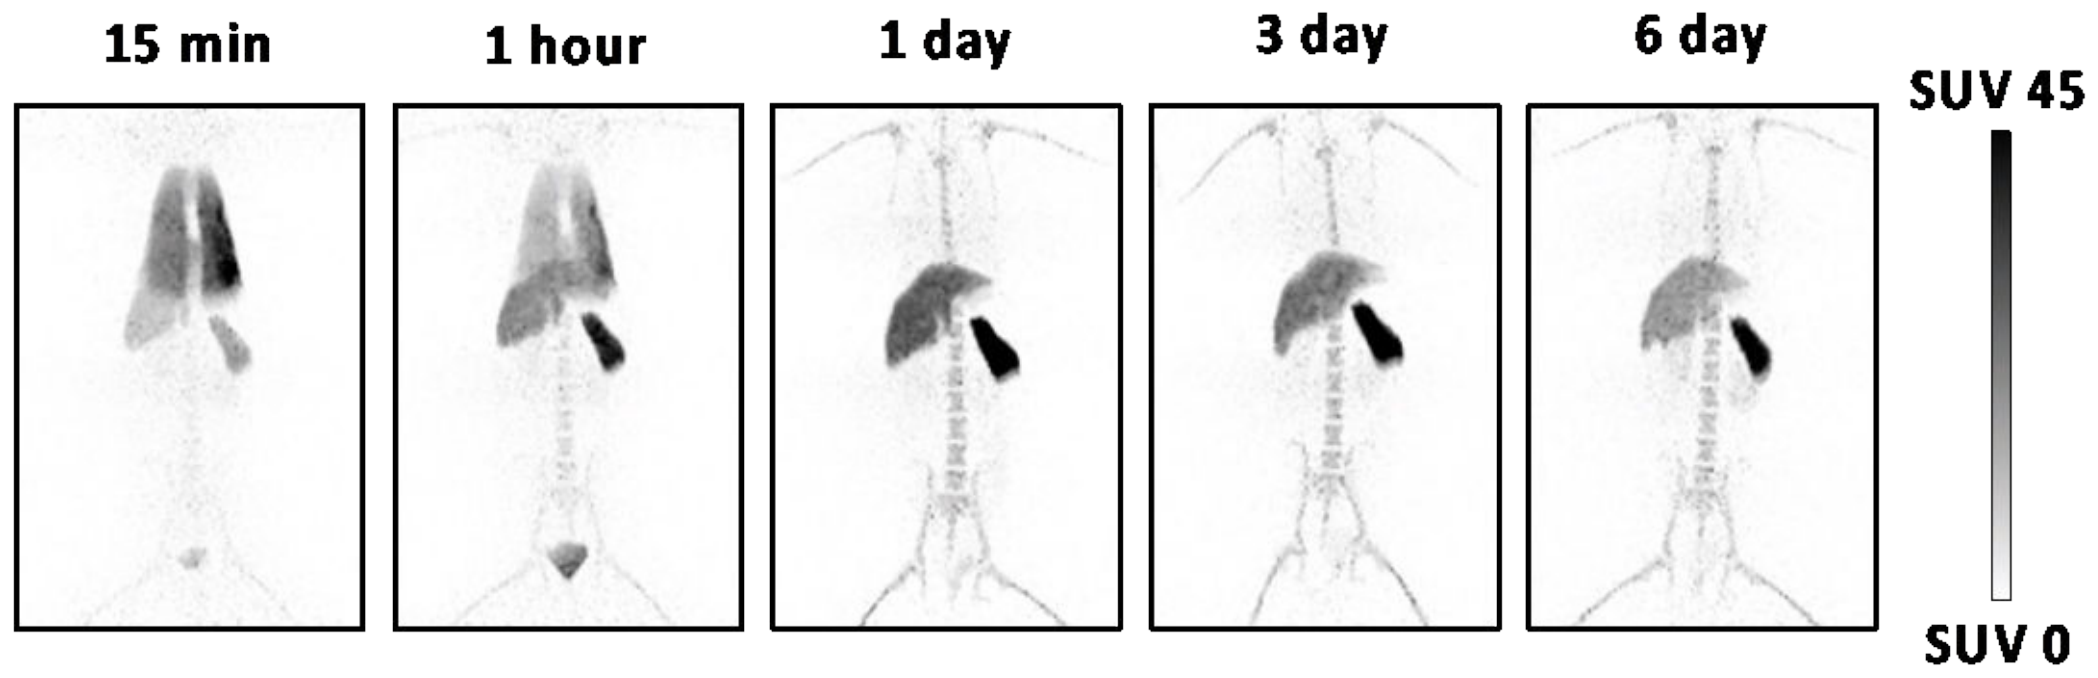
15 min
1 hour
1 day
3 day
6 day
SUV 45
SUV 0